139

Final Analysis of COMET-1: Cabozantinib versus Prednisone in Metastatic Castration-Resistant Prostate Cancer (mCRPC) Patients Previously Treated With Docetaxel and Abiraterone and/or Enzalutamide

Smith MR, de Bono JS, Sternberg CN, Le Moulec S, Oudard S, De Giorgi U, Krainer M, Bergman AM, Hölzer W, De Wit R, Bögemann M, Saad F, Cruciani G, Thiery-Vuillemin A, Feyerabend S, Miller K, Ramies DA, Hessel C, Weitzman AL, and Fizazi K

INTRODUCTION

Cabozantinib is an Oral Inhibitor of Tyrosine Kinases Including MET and VEGFR2

- Antiangiogenic and antitumor effects in a wide range of preclinical tumor models[1-2]
- Blocks progression of prostate tumor xenografts in soft tissue and/or bone[3-5]
- Effects on the bone microenvironment[3-6]
 - Reduction in osteoclasts
 - Biphasic effects (↑→↓) on osteoblasts
 - Alters bone remodeling (↑volume) in mice

Figure 1. ARCaP-M CRPC xenografts in mice

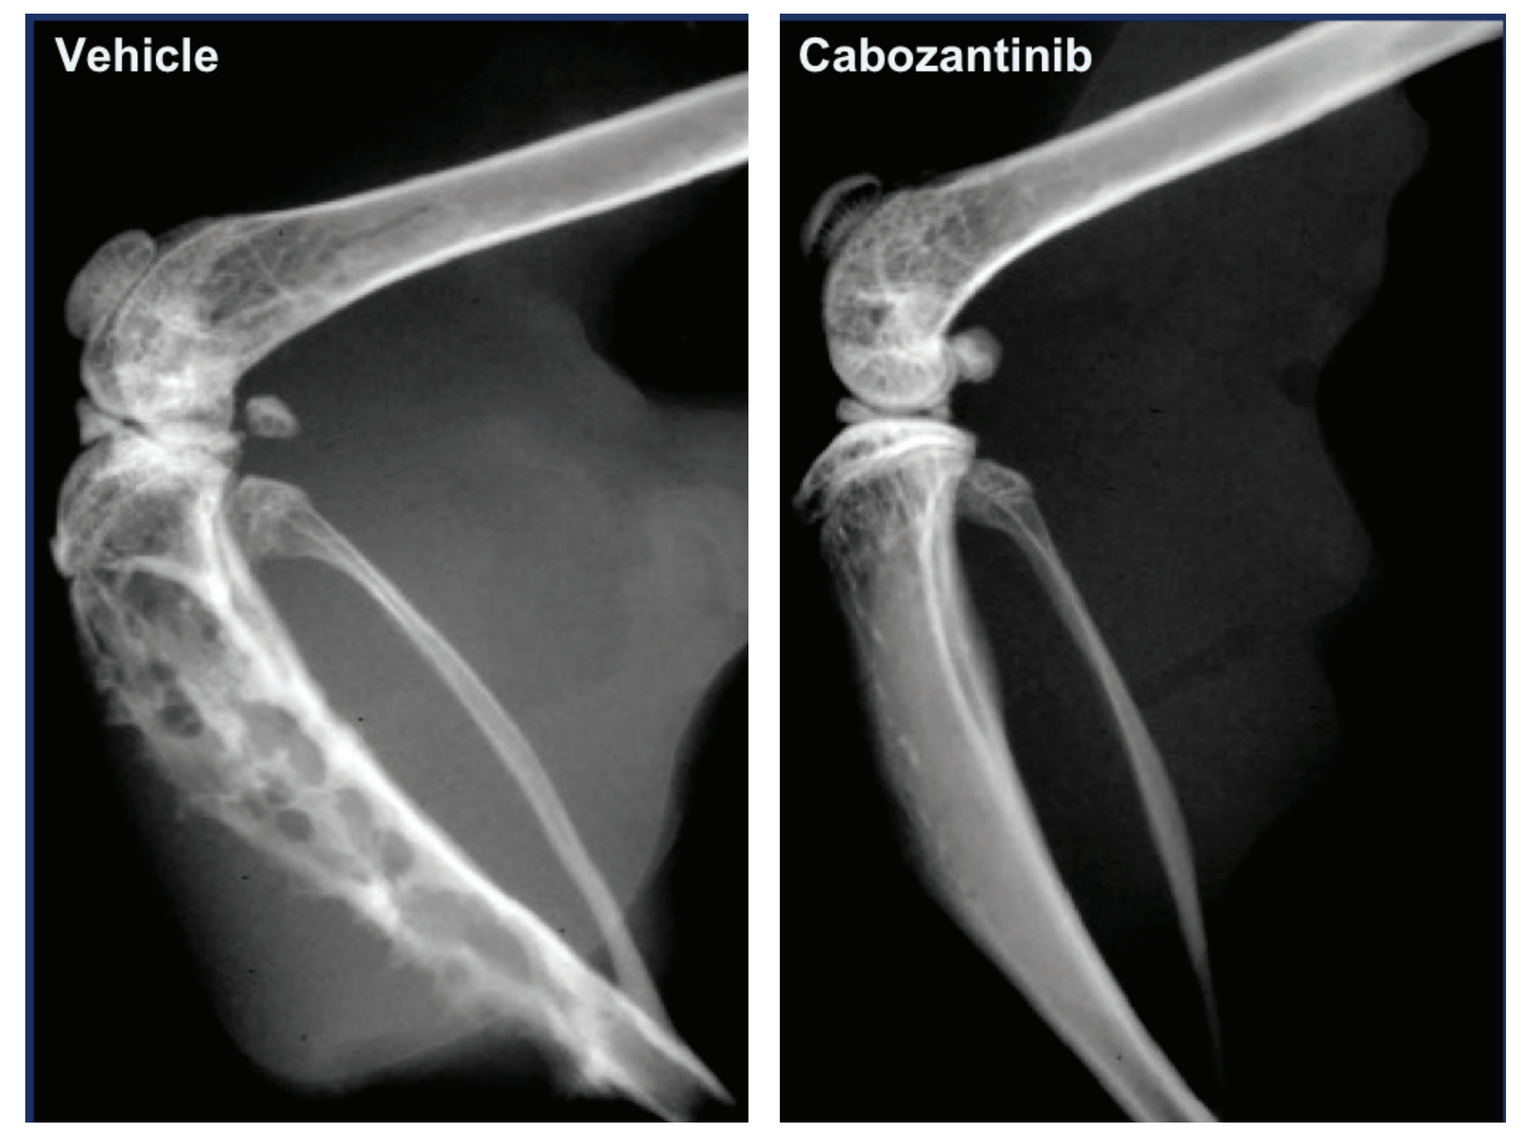
Vehicle
Cabozantinib

Schimmoller et al., *Mol Cancer Ther* (2011) 10: A233

Phase 2 Observations in >300 mCRPC Patients

- Bone effects: high rate of bone scan resolution and reduction of bone biomarkers[7,8]
- Soft tissue effects: objective response and significant progression-free survival benefit[7]
- Improvement in pain and reduction of narcotics[9]
- Reduction in circulating tumor cells[8]
- Activity observed in patients heavily pretreated with docetaxel, abiraterone, and/or enzalutamide[8,9]
- Lowest effective dose: 40 mg/day[10]

Figure 2. Bone scan images

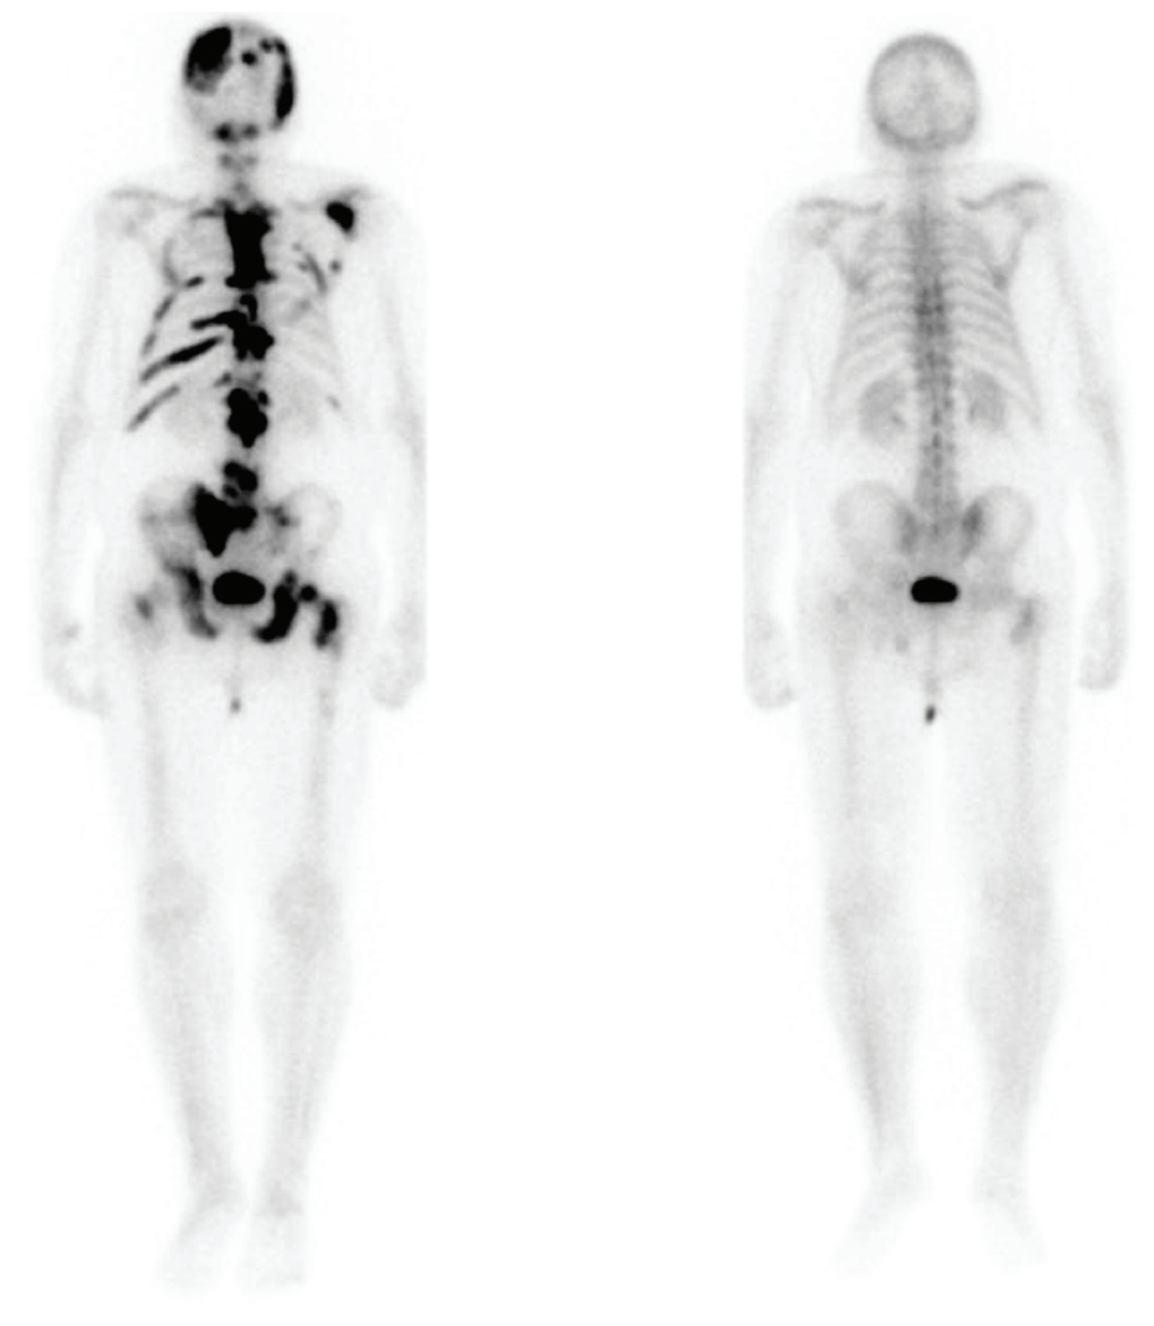

Baseline Follow-up

COMET-1 STUDY DESIGN

Figure 3. International Phase 3 Trial

mCRPC N = 960
- Post-progression w/ docetaxel
- Post-progression w/ abiraterone and/or enzalutamide
- No limit to prior treatments
- Bone metastases (with or without severe pain)

Arm A (n = 640): Cabozantinib 60 mg qd + Placebo

Arm B (n = 320): Prednisone 5 mg bid + Placebo

METHODS

Statistical Methods & Assumptions

- Primary endpoint: overall survival
- Secondary endpoints: bone scan response, skeletal-related events, circulating tumor cells, and bone biomarkers
- Stratification factors
 - Prior cabazitaxel (yes/no)
 - Baseline pain severity (worst pain by Brief Pain Inventory [BPI] Item 3; < 4 versus ≥ 4)
 - ECOG performance status (0-1 versus 2)
- Sample size: 960 (640 cabozantinib; 320 prednisone)
- Assumed OS of 9.2 months for cabozantinib arm; 7 months for prednisone arm
 - COU-AA-301: time from end of treatment to median OS = 7 months
- 90% power to detect 25% reduction in risk of death (HR=0.75), 2-sided type 1 error = 0.05
 - 578 events required for primary analysis
 - Single interim analysis planned after 386 events (67% of events)
 - Sequential testing and alpha spending function used to control type 1 error

RESULTS

Figure 4. Patient Disposition

Randomized (2:1) N = 1028

	Cabozantinib N = 682	Prednisone N = 346
Continuing treatment	9%	9%
Discontinued treatment	**91%**	**95%**
Progressive Disease	**19%**	**37%**
Adverse Event	**33%**	**12%**
Clinical deterioration	33%	38%
Subject withdrawal	3%	4%
Investigator decision	1%	2%
Did not receive treatment	0.3%	0.9%
Other	0.1%	0.6%
Included in ITT analysis	n = 682	n = 346
Included in safety analysis	n = 680	n = 343
Duration of exposure (mo)	4.6	2.8

ITT, intent-to-treat

RESULTS

Table 1. Baseline Characteristics

	Cabozantinib (N=682)	Prednisone (N=346)
Median age (min; max)	69.5 (35, 87)	69.0 (43, 89)
ECOG PS 2	11%	12%
Prior cabazitaxel	38%	38%
Prior abiraterone	92%	92%
Prior enzalutamide	25%	26%
Prior abiraterone + enzalutamide	16%	19%
Visceral disease (liver or lung)	20%	17%
BPI worst pain ≥ 4	42%	43%
Narcotics within past 24 hours	67%	66%
Bisphosphonates/denosumab	48%	49%

Figure 5. Overall Survival

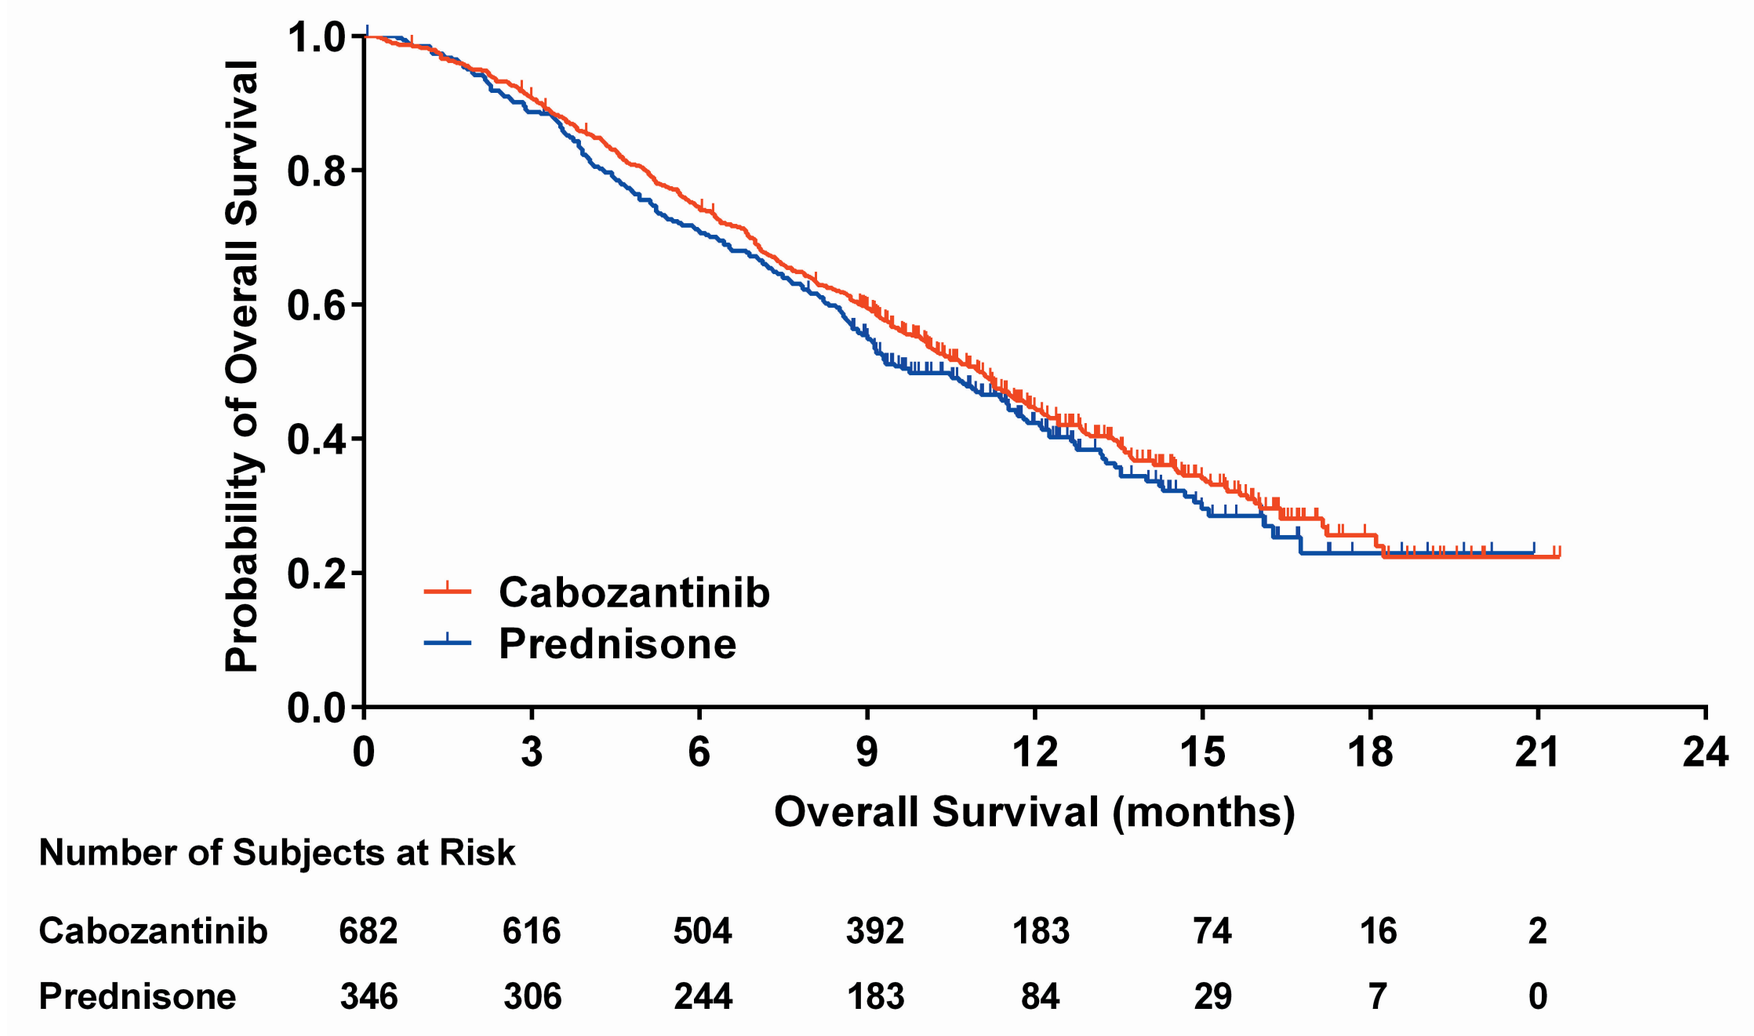
Probability of Overall Survival
1.0
0.8
0.6
0.4
0.2
0.0
Cabozantinib
Prednisone
0 3 6 9 12 15 18 21 24
Overall Survival (months)
Number of Subjects at Risk
Cabozantinib 682 616 504 392 183 74 16 2
Prednisone 346 306 244 183 84 29 7 0

	N (Events)	Median (mo) (95% CI)	Stratified HR (95% CI)	Stratified LR p-value
Cabozantinib	682 (403)	11.0 (10.1, 11.6)	0.90 (0.76, 1.06)	0.213
Prednisone	346 (211)	9.8 (9.0, 11.5)		

CI, confidence interval; HR, hazard ratio; LR, log-rank test

Figure 6. Subgroup Analyses

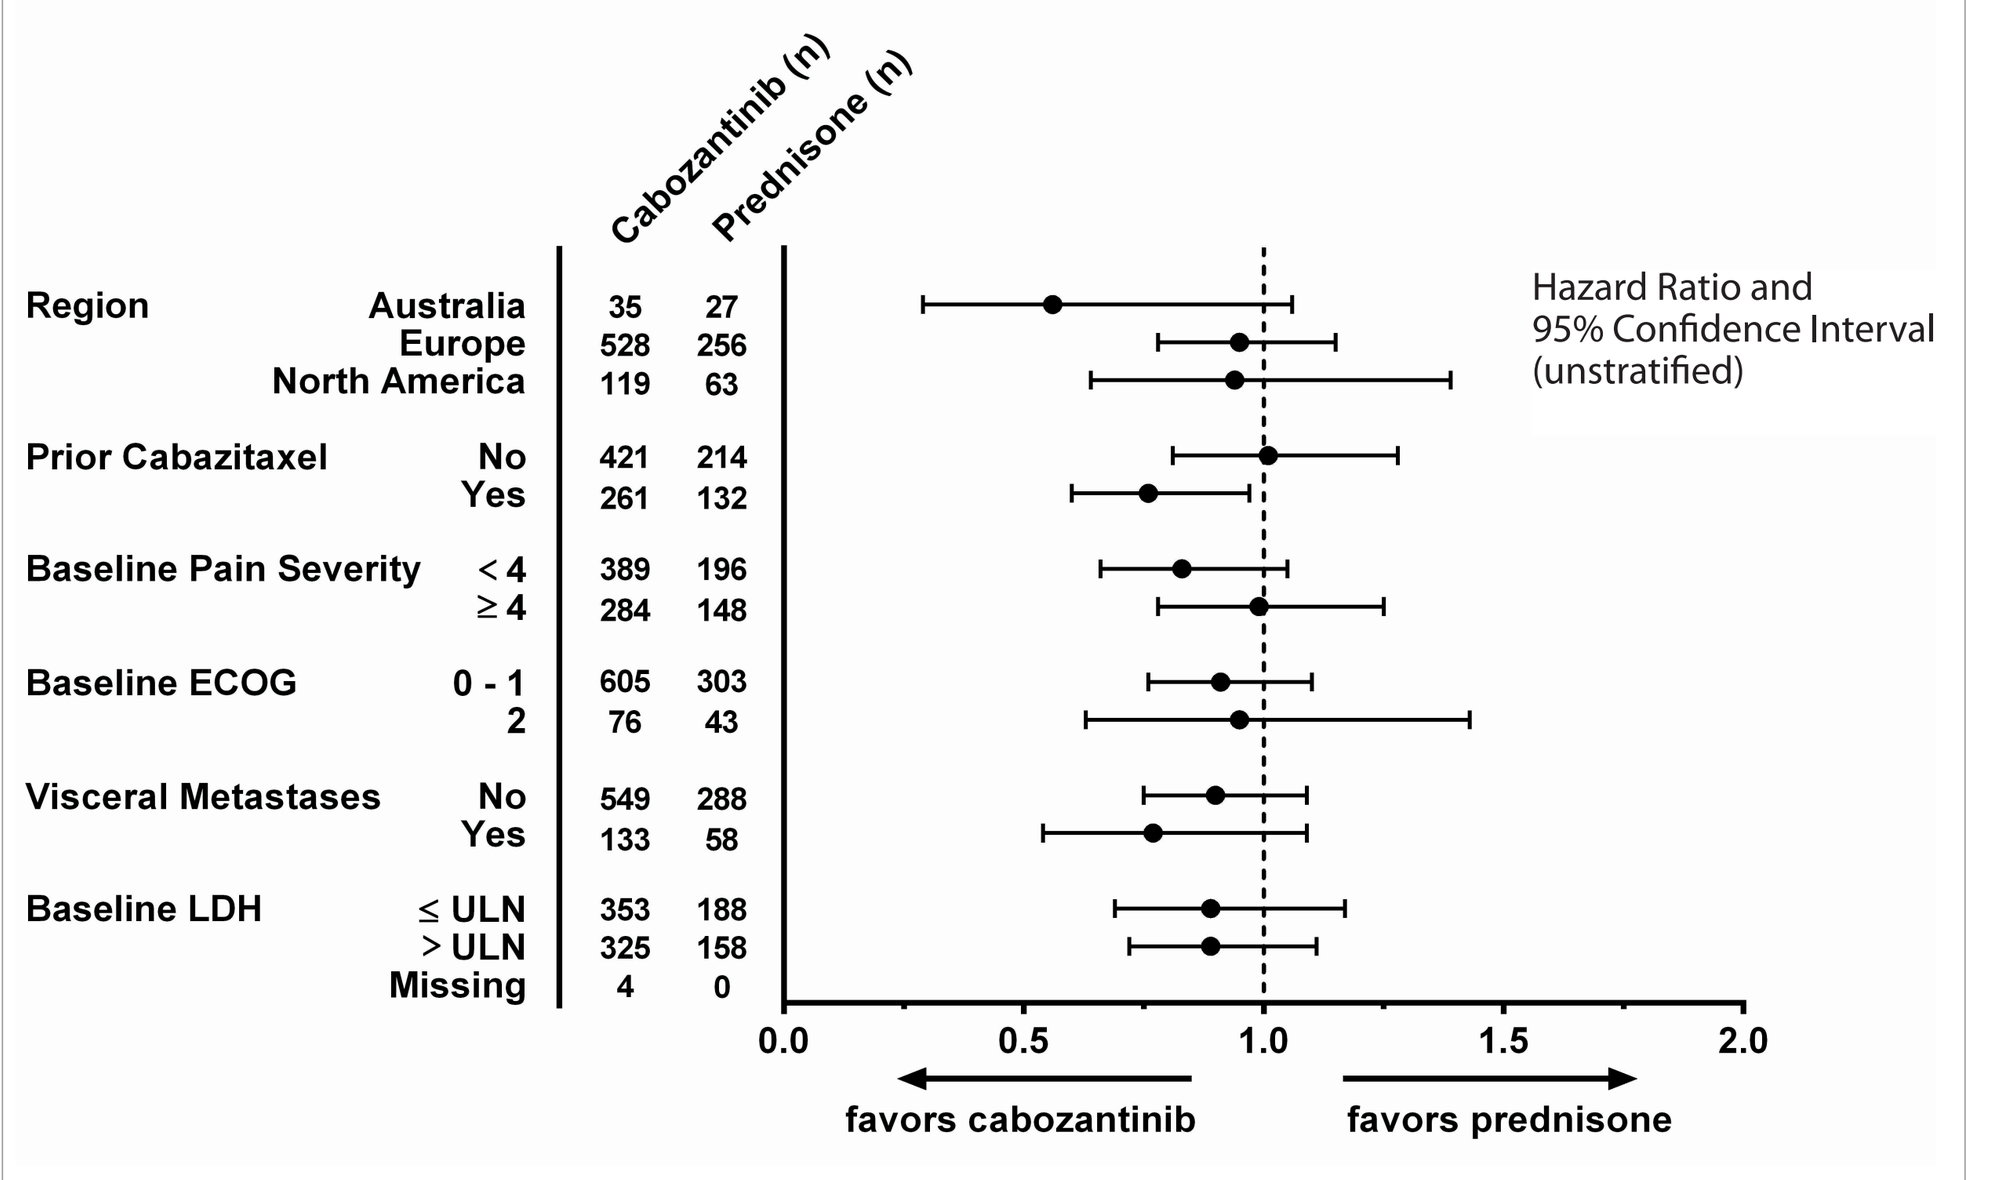
Cabozantinib (n)
Prednisone (n)
Region Australia 35 27
Europe 528 256
North America 119 63
Prior Cabazitaxel No 421 214
Yes 261 132
Baseline Pain Severity < 4 389 196
≥ 4 284 148
Baseline ECOG 0 - 1 605 303
2 76 43
Visceral Metastases No 549 288
Yes 133 58
Baseline LDH ≤ ULN 353 188
> ULN 325 158
Missing 4 0
Hazard Ratio and 95% Confidence Interval (unstratified)
0.0 0.5 1.0 1.5 2.0
favors cabozantinib
favors prednisone

LDH, lactate dehydrogenase; ULN, upper limit of normal

Table 2. Subsequent Anti-Cancer Therapy (ITT Population)

	Cabozantinib (N = 682)	Prednisone (N = 346)
Patients with any therapies*	55%	68%
Docetaxel	2.5%	8.7%
Cabazitaxel	14%	23%
Enzalutamide	20%	22%
Abiraterone	5.4%	7.5%
Radium-223	6.2%	5.5%
External Beam Radiation Therapy	12%	20%

* Patients may have had more than one subsequent therapy
ITT, intent-to-treat; WHO DRUG version March 2014 was used for coding

Table 3. Bone Scan Response at Week 12 per Independent Central Review (ITT Population)

	Cabozantinib (N = 682)	Prednisone (N = 348)
Bone scan response rate*	**42%**	**3%**
95% confidence interval	(38%, 46%)	(1%, 5%)
Stratified CMH test p-value	<0.001	
Duration of response (months)	5.8	1.8
95% CI (months)	(5.5, 8.3)	(0.6, not estimable)
Stable bone scan	**20%**	**29%**
Progressive bone scan	**10%**	**37%**
Not evaluable	**28%**	**31%**

* Bone scan response = ≥30% reduction in bone scan lesion area by central review (Brown et al. *Nucl Med Commun* 2012)
CMH, Cochran-Mantel-Haenszel; IRF, independent radiology review facility; ITT, intent to treat.

Figure 7. Progression-Free Survival (Investigator Assessed)

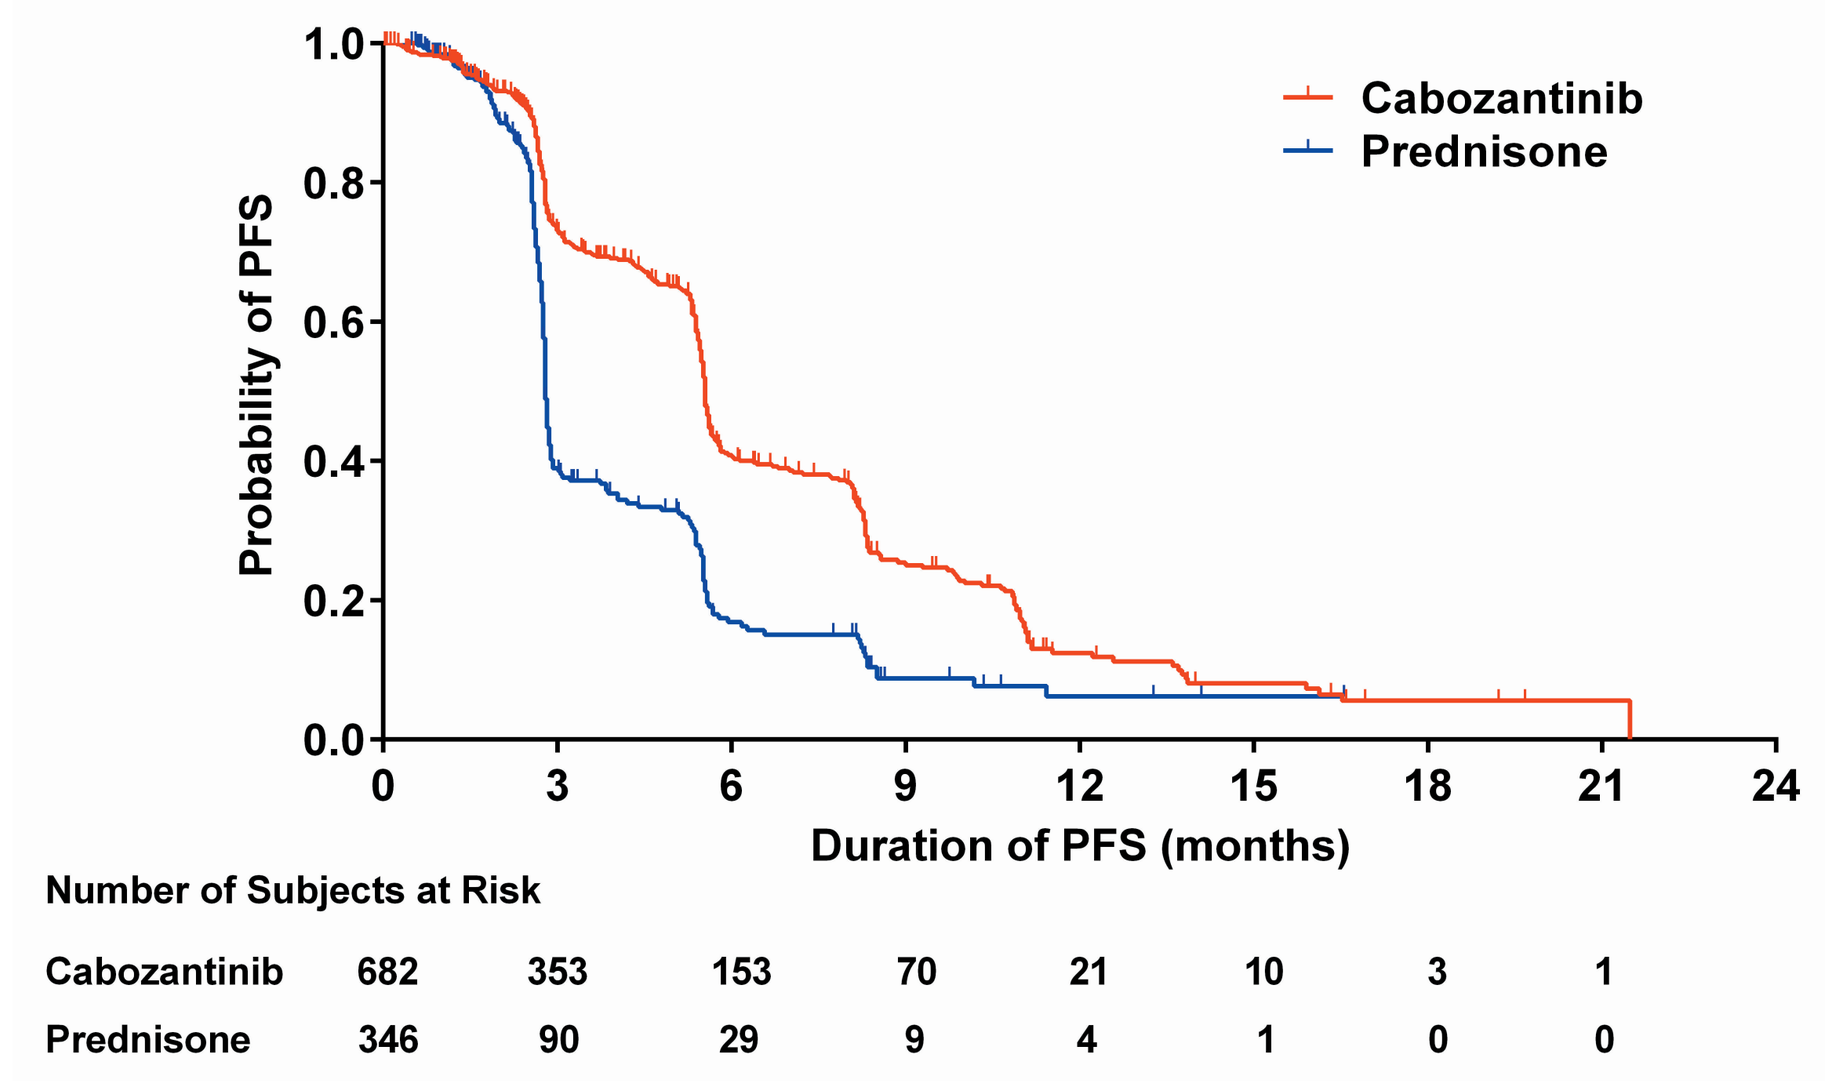
Probability of PFS
1.0
0.8
0.6
0.4
0.2
0.0
Cabozantinib
Prednisone
0 3 6 9 12 15 18 21 24
Duration of PFS (months)
Number of Subjects at Risk
Cabozantinib 682 353 153 70 21 10 3 1
Prednisone 346 90 29 9 4 1 0 0

	N (Events)	Median (mo) (95% CI)	Stratified HR (95% CI)	Stratified LR p-value
Cabozantinib	682 (375)	5.6 (5.5, 5.6)	0.48 (0.40, 0.57)	<0.001
Prednisone	346 (220)	2.8 (2.8, 2.9)		

CI, confidence interval; HR, hazard ratio; ITT, intent-to-treat; LR, log-rank test; PFS, progression-free survival

Figure 8. Time to First SRE on Study

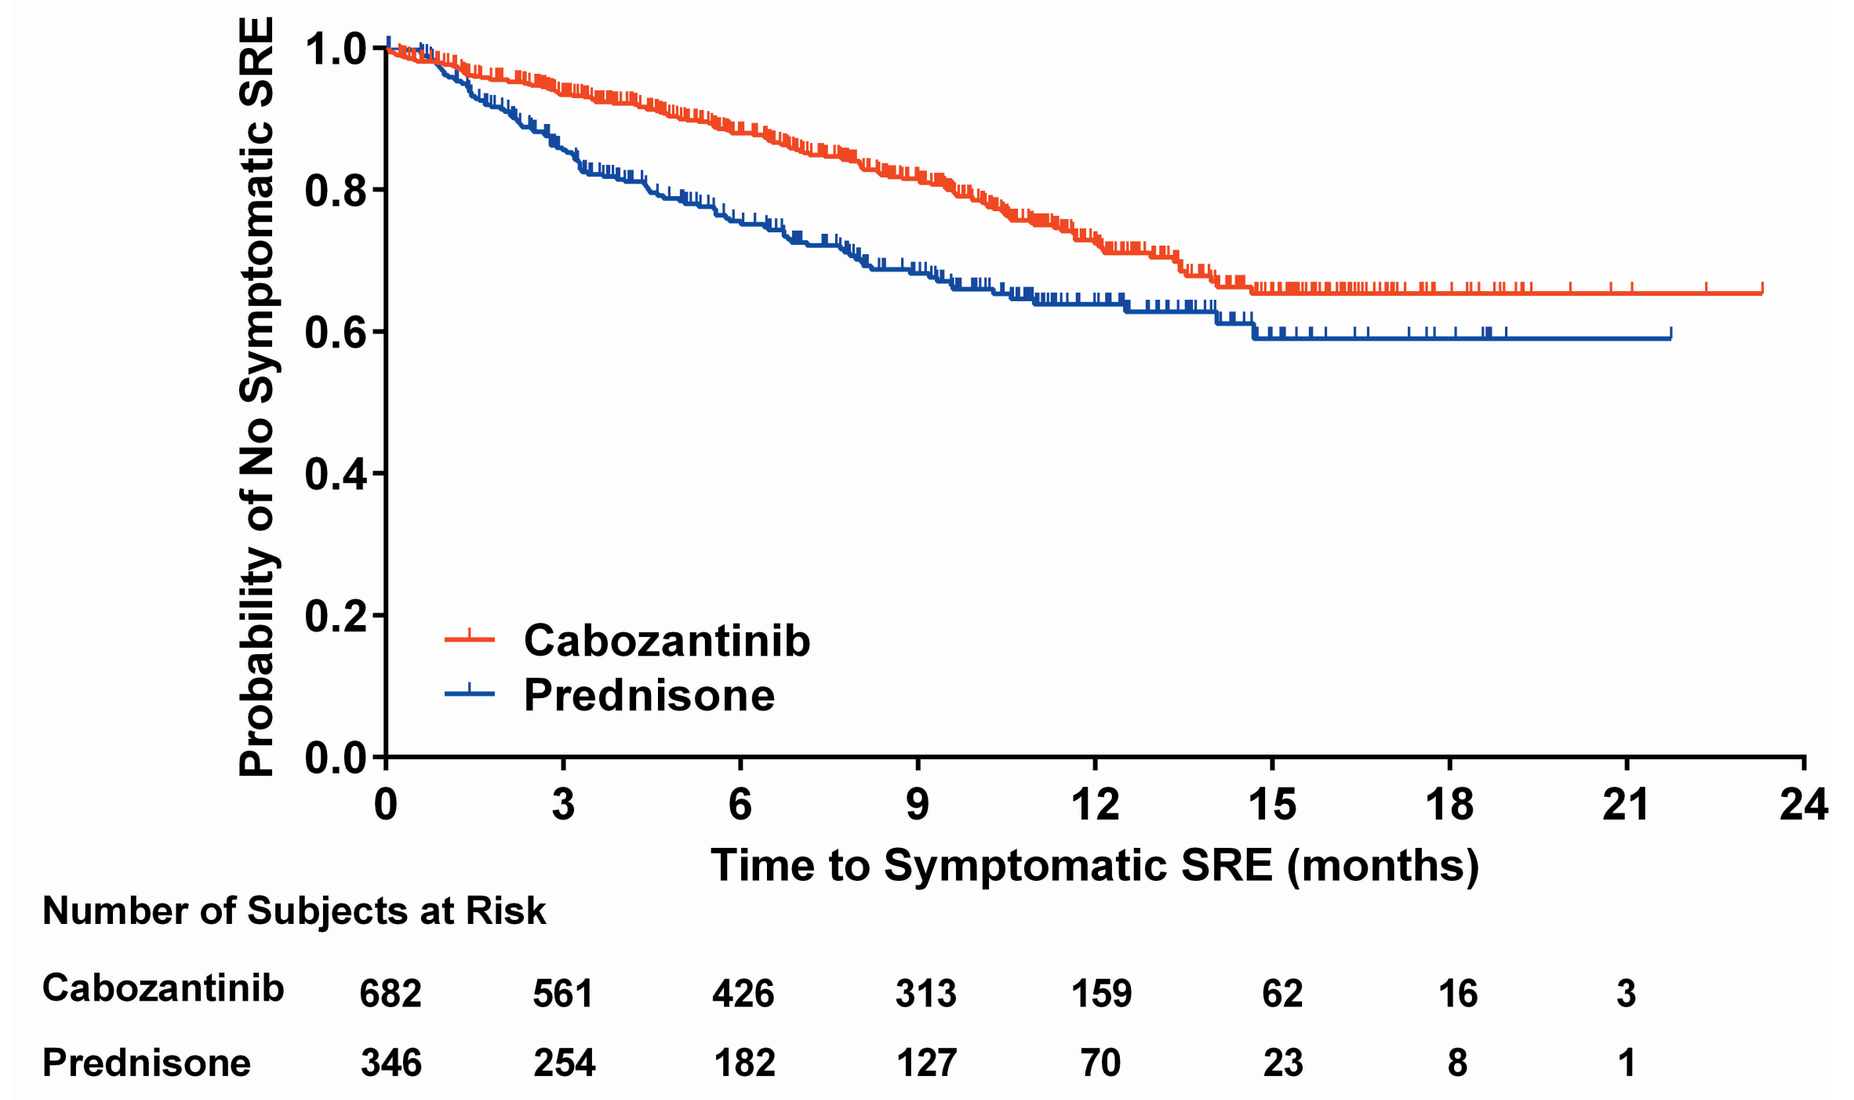
Probability of No Symptomatic SRE
1.0
0.8
0.6
0.4
0.2
0.0
Cabozantinib
Prednisone
0 3 6 9 12 15 18 21 24
Time to Symptomatic SRE (months)
Number of Subjects at Risk
Cabozantinib 682 561 426 313 159 62 16 3
Prednisone 346 254 182 127 70 23 8 1

	N (Events)	Median (mo) (95% CI)	Stratified HR (95% CI)	Stratified LR p-value
Cabozantinib	682 (139)	NE (NE, NE)	0.62 (0.48, 0.81)	<0.001
Prednisone	346 (100)	NE (NE, NE)		

SRE rate: 14% cabozantinib vs. 21% prednisone

SRE, skeletal-related event; CI, confidence interval; HR, hazard ratio; NE, not estimable; LR, log-rank test

Additional Endpoints

- CTCs: conversion from ≥ 5 at baseline to < 5 on treatment
 - 33% cabozantinib vs. 6% prednisone
- Bone biomarkers (best change from baseline)
 - BSAP: ↓10% cabozantinib vs. ↑11% prednisone
 - NTx: ↓24% cabozantinib vs. 0% prednisone
 - CTx: ↓12% cabozantinib vs. 0% prednisone

CTCs, circulating tumor cells; BSAP, bone-specific alkaline phosphatase; CTx (NTx), C-terminal (N-terminal) cross-linked telopeptides of type I collagen

SAFETY

Table 4. Safety: Exposure and Dose Reductions

	Cabozantinib (N = 681)	Prednisone (N = 342)[a]
Median duration of exposure – months (95% CI)[b]	4.6 (0.14, 21.8)	2.8 (0.46, 16.4)
Median percent dose intensity of cabozantinib / placebo[c]	75%	100%
Experienced ≥ 1 dose reduction	60%	13%

a One patient in the placebo arm who received cabozantinib was removed from the placebo safety population and added to the cabozantinib safety population
b Duration of exposure = (Date of decision to discontinue study treatment – Date of first dose + 1) / 7
c Percent dose intensity of cabozantinib = 100 x (Average daily dose mg/day) / (60 mg/day)

Table 5. Safety Overview

	Cabozantinib (N = 681)	Prednisone (N = 342)
Adverse events – any grade	100%	97%
Adverse events – grade 3/4	71%	56%
Serious adverse events	62%	53%
Discontinued due to adverse event	33%	12%
Died ≤ 30 days of last dose	16%	13%

Table 6. Summary of Frequent Grade 3/4 Adverse Events
Safety Population (>5% of Patients in Cabozantinib Arm)

Adverse Event	Cabozantinib (N = 681)	Prednisone (N = 342)
Hypertension	20%	7.0%
Fatigue	17%	8.8%
Anaemia	16%	18%
Asthenia	12%	6.1%
Decreased appetite	8.1%	2.0%
General physical health deterioration	7.8%	5.8%
Diarrhoea	7.3%	1.5%
Nausea	6.9%	2.0%
Hand-foot syndrome	5.7%	0
Vomiting	5.6%	2.3%
Bone pain	5.1%	6.7%

CONCLUSIONS

- Cabozantinib did not significantly improve OS (HR 0.90, p=0.21) in men with mCRPC and disease progression despite prior docetaxel and abiraterone and/or enzalutamide
- Subgroup analyses suggest larger OS improvement in men with visceral metastases or prior cabazitaxel
- Cabozantinib was associated with significant improvements in bone scan response, PFS, time to first SRE, CTC conversion, and bone biomarkers
- Observed safety profile is consistent with prior studies

REFERENCES

1. Yakes, *Mol Cancer Ther*, 2011; 2. Sennino, *Cancer Discov*, 2012; 3. Nguyen, *PLoS One*, 2013 ; 4. Dai, *Clin Cancer Res*, 2014; 5. Graham, *J Natl Cancer Inst*, 2014; 6. Stern, *J Cell Biochem*, 2014; 7. D. Smith, et al., *J Clin Oncol*, 2013; 8. M. Smith, et al., *J Clin Oncol*, 2014; 9. E. Basch, et al., *Eur Urol*, 2015; 10. R. Lee, et al., *Clin Cancer Res*, 2013

ACKNOWLEDGEMENTS

We are grateful to all of the COMET-1 patients and their caregivers, and to all the principal investigators who participated in this study.

Graphic design assistance was provided by Price Watkins Media, and funded by Exelixis.

Presented at the Genitourinary Cancers Symposium; February 26–28, 2015; Orlando, Florida